Rapid Aggregate Reduction of Bi-Specific Antibody Model by Filtration

Joy Adiletta[1], Roger Alsop[2,3] Carl Breuning[2] Larissa Pecore[1] and Amanda Mak[1]

[1]SystImmune, Inc. 15318 NE 95th St, Redmond, WA, 98052

[2]Sartorius Stedim Biotech North America Bohemia NY

[3]roger.alsop@sartorius-stedim.com

Abstract

Removal of protein aggregates from biologic drug products is critical due to their potential to increase antigenic response and the associated negative impact on the patients. Aggregates may be formed during upstream or downstream operations when suboptimal purification parameters are selected.

Once formed, aggregate removal and/or reduction becomes challenging for the downstream purification process development team. Chromatography is the most traditional purification technology used for aggregate removal; however, chromatography methods could actually increase aggregate formation as the product is exposed to harsh conditions such as low pH or high salt. Column chromatography is also time consuming, requiring several buffers, method development, and costly technology such as purification skids and columns.

SystImmune and Sartorius are exploring an alternate technique exploiting the Virosart® Max filter. The Virosart® Max is an optimized triple-layer polyamide pre-filter specifically designed to remove aggregates and protect the expensive downstream virus removal filter. The Virosart® Max will be operated in a simple flow through mode to demonstrate aggregate reduction in a bi-specific antibody model.

This robust filter-based aggregate removal strategy would offer simple buffer preparation, minimal process development, and the option to replace costly chromatographic skids with inexpensive pumping equipment.

Introduction:

The Virosart® Max was specifically designed as a pre-filter to mitigate aggregates during the final virus retentive filtration step. By removing the most challenging agents for the viral filter, aggregates and/or small hydrophobic molecules, the Virosart® Max will improve throughput through the virus retention filter. Virosart® Max binds aggregates very efficiently through hydrophobic interactions, independently of process conditions such as conductivity. The triple layer polyamid membrane material provides high adsorptive capacities optimized for this process step.

Materials and Method

- Product: A Bispecific Ab 4 mg/mL; aggregate 10 % HMW species
 - Virosart® Max filter; 5 cm^2 part no: 54AMI----------B
 - 0.1 µm Minisart filter; 5 cm^2 part no: 16553----------K
- Buffers :
 - 20 mM Sodium phosphate, pH 7.0
 - 20 mM TRIS pH 7.8
 - 20 mM Sodium phosphate, pH 6.5
 - 20 mM TRIS / 2 M NaCl, pH 7.8
 - 20 mM Sodium phophate/ 2 M NaCl pH 7.0
 - Deionized Water

AKTA AVANT

The Virosart® MAX was installed onto AKTA Avant and the system was in programmed method mode.

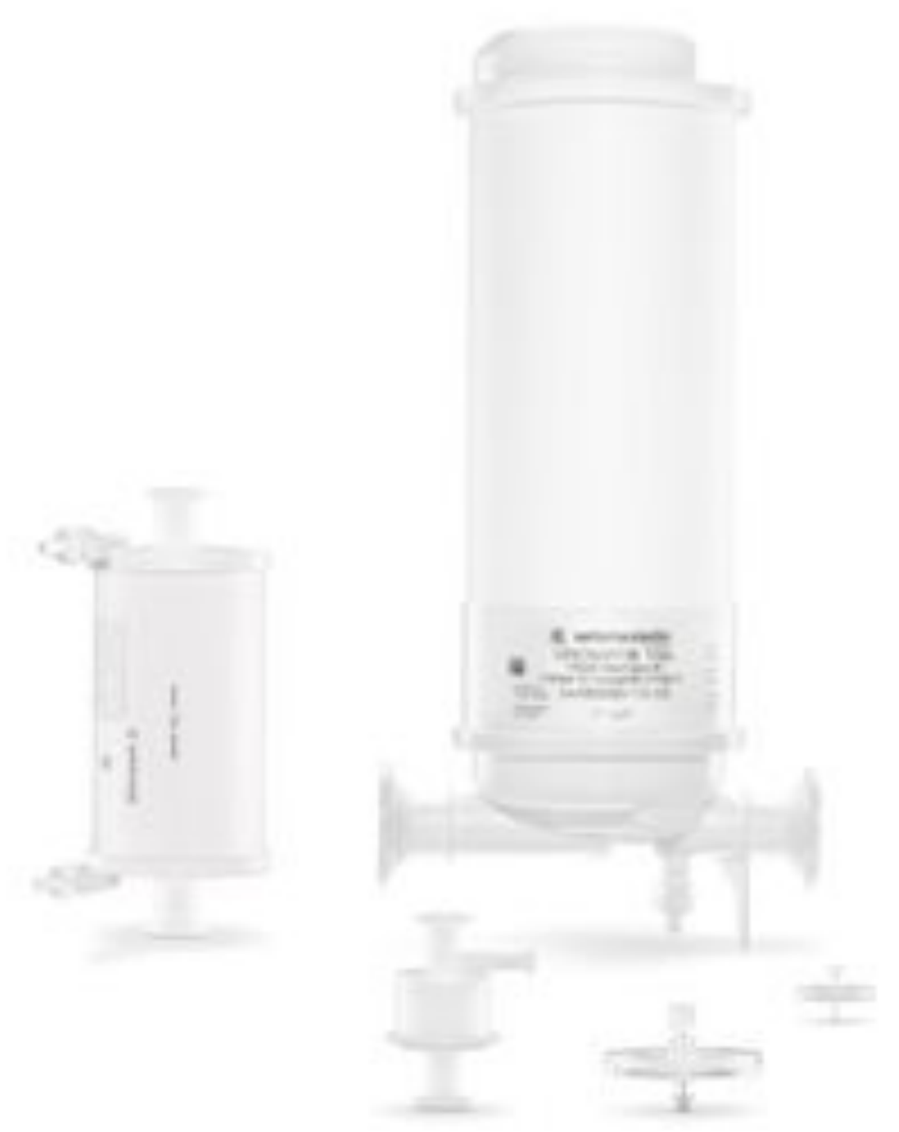

Figure 1: Virosart Max

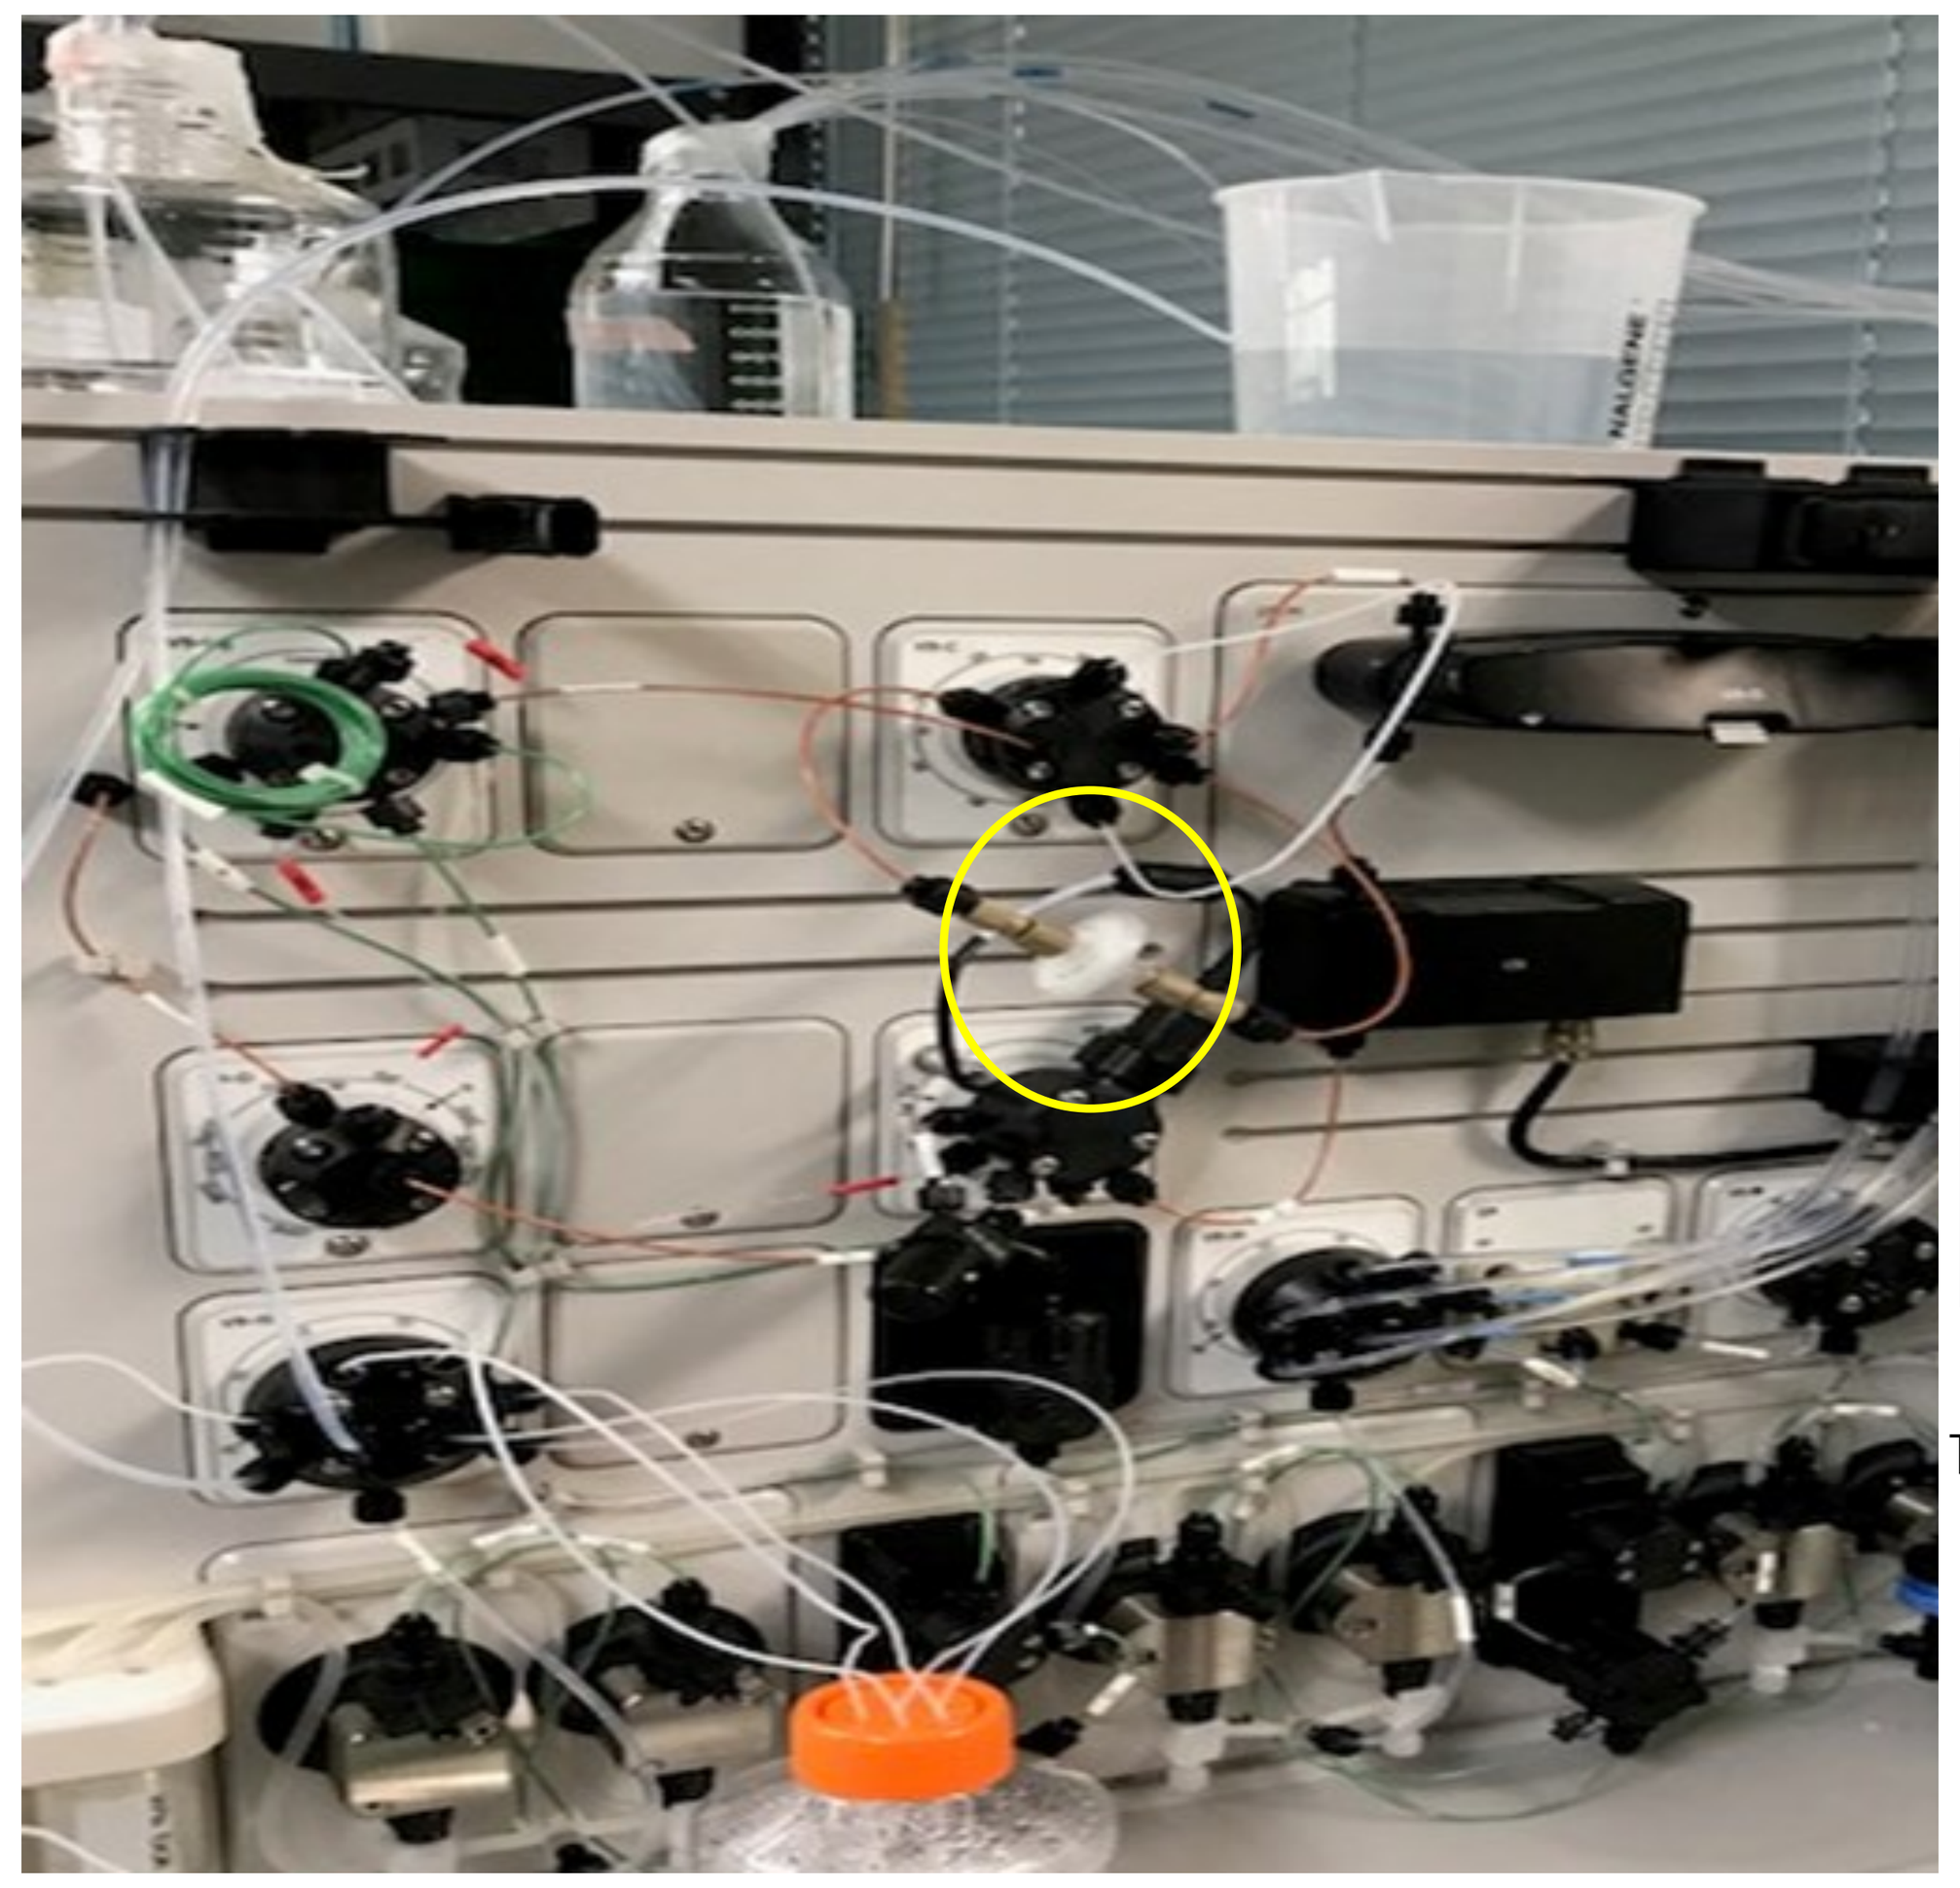

Figure 2: Virosart Max Installed on AKTA Avant

Table 1 Protocol

Ran in constant flow mode (5 mL/min)
1. 20 mL DI Water
2. 20 mL buffer
3. Load (fraction collected)
4. 20 mL Buffer wash

Table 2: Run Parameters

Run	Load	Volume (mL)	Buffer	Fraction size (mL)
1	neat	20	20 mM Sodium phophate, pH 7.0	1
2	neat	20	20 mM TRIS, pH 7.8	1
3	neat	20	20 mM Sodium phophate, pH 6.5	1
4	Dil 1:1	40	20 mM TRIS, 2 M NaCL pH 7.8	2
5	Dil 1:1	40	20 mM Sodium phosphate, 2 M NaCl pH 7.0	2
6	Neat	20	20 mM Sodium phosphate, pH 7.0 (0.1 µm)	1

The collected fractions were submitted for SEC-HPLC and A280 (protein quantitative) analytics

Results and Discussion

Chromatograms

Results from Table 2 for the neat (undiluted) sample only as diluted samples were below levels of detection for SEC-HPLC assay .

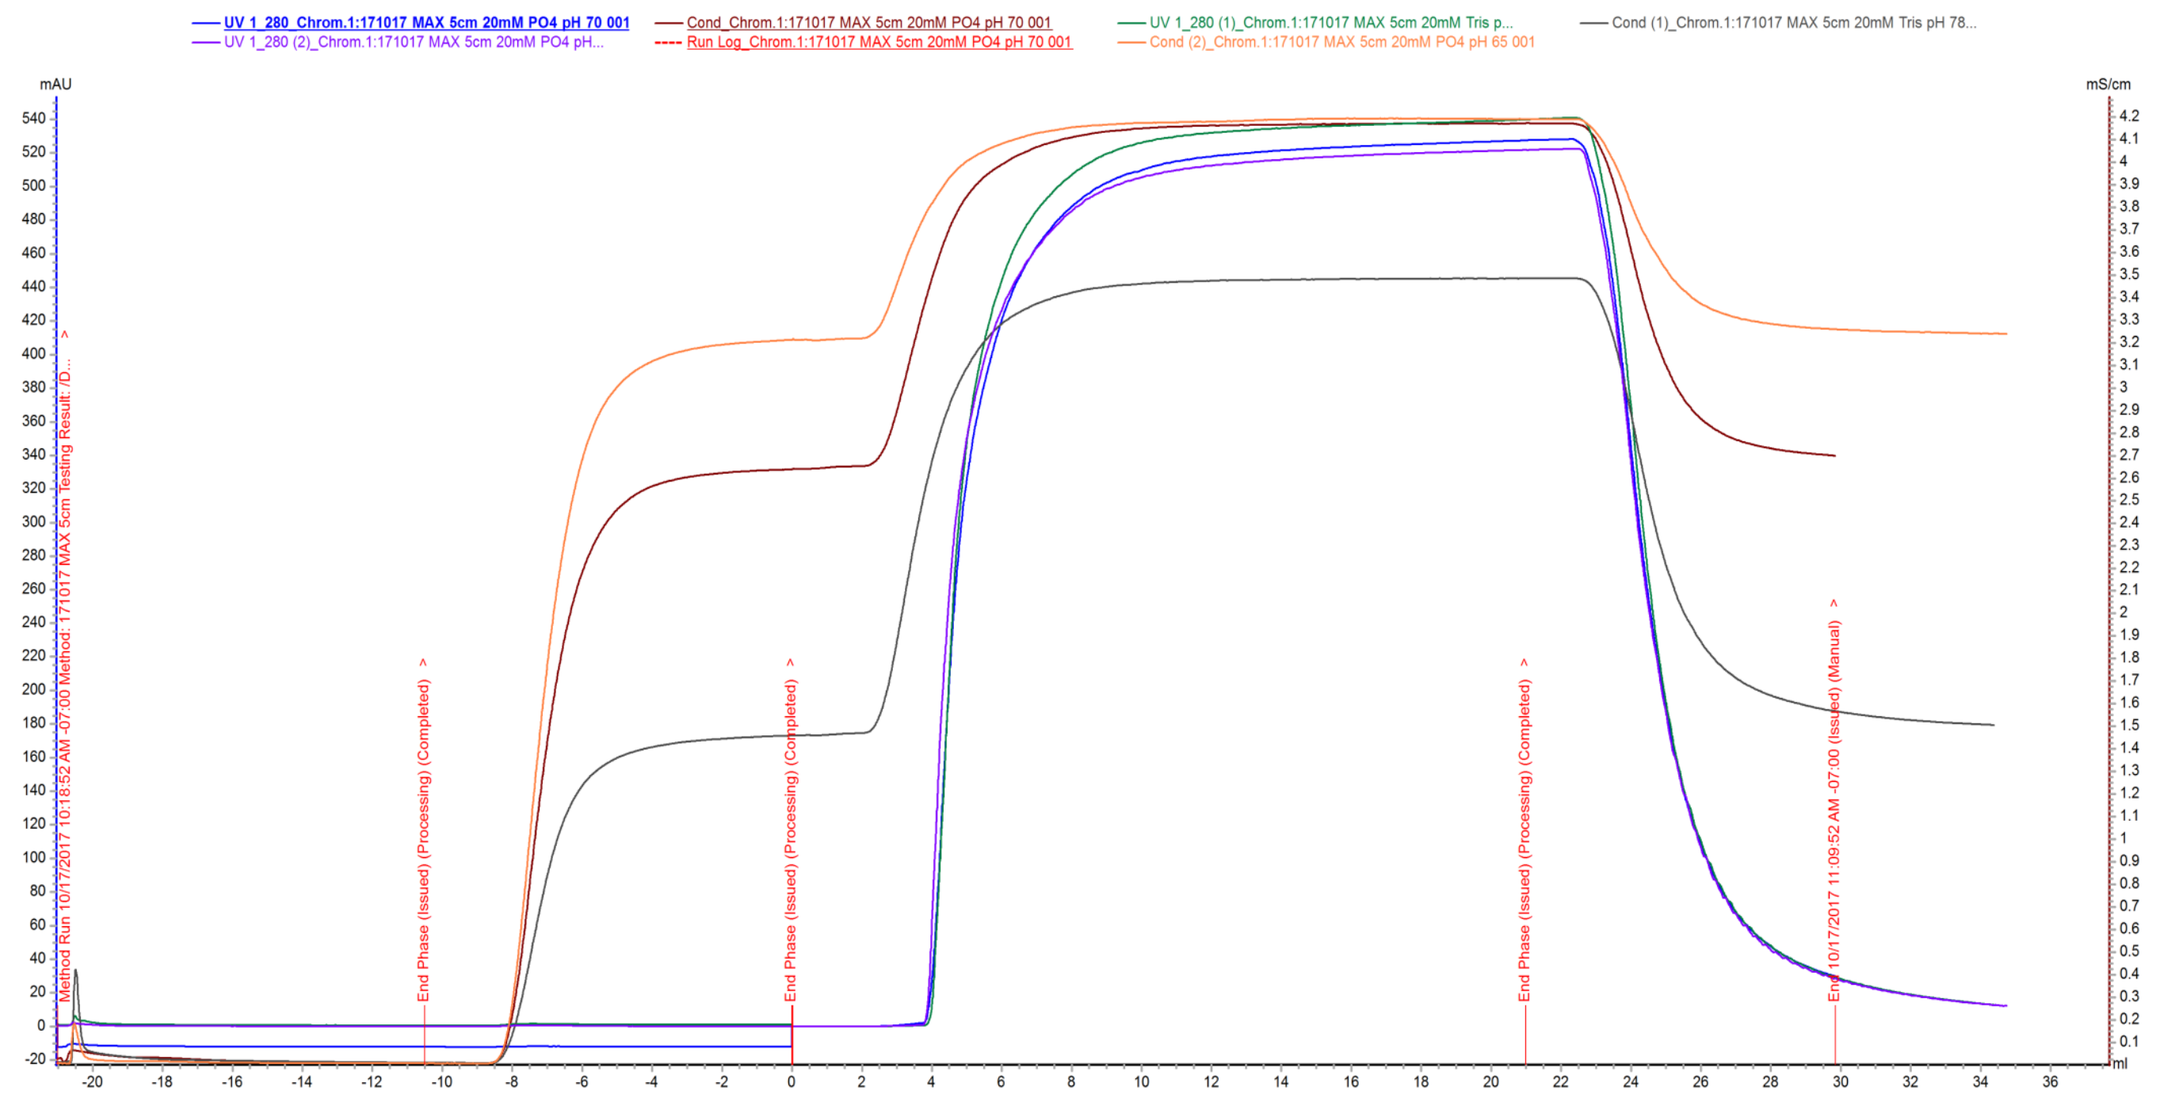

Figure 3A: Overlays pH 5.6/7.0/7.8 non-salt conditions

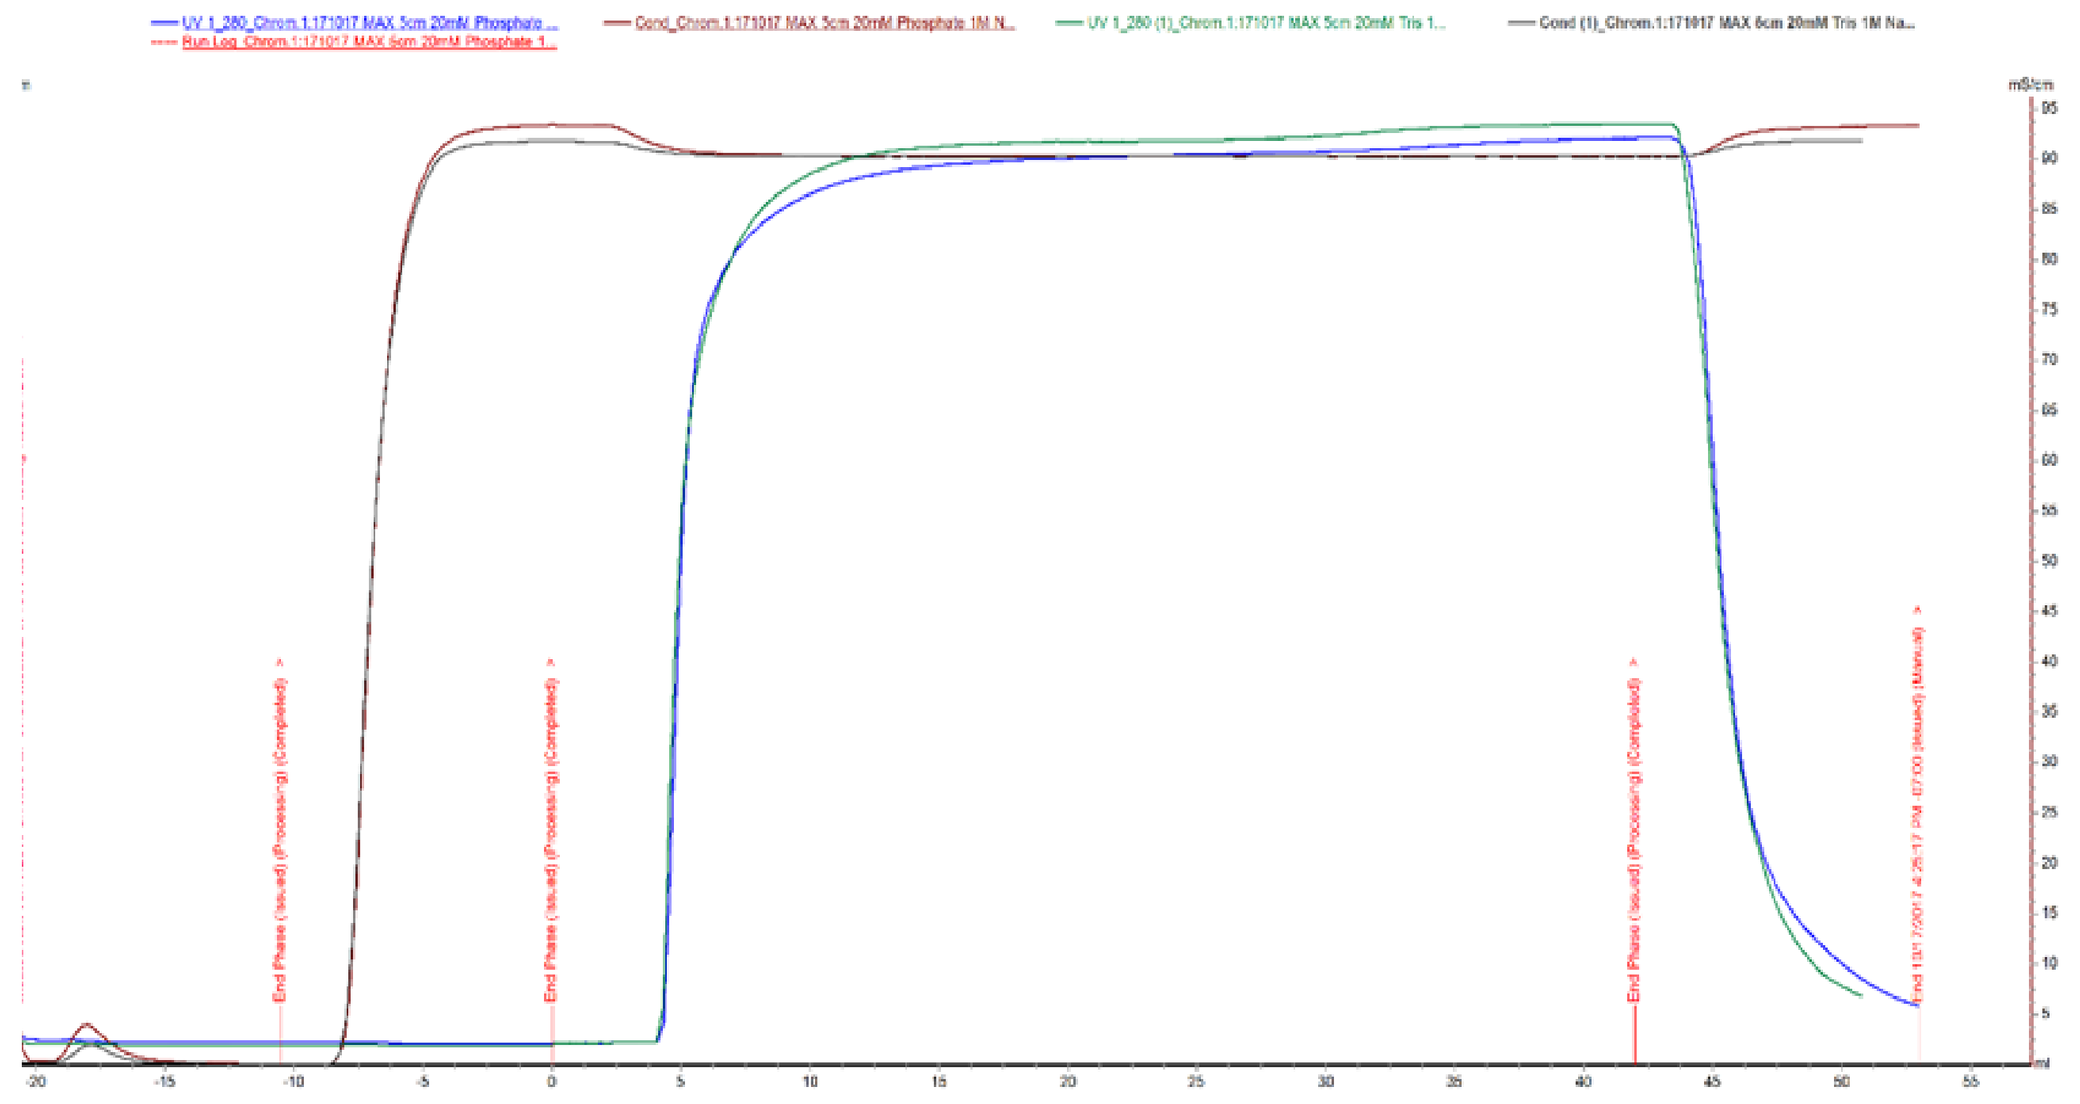

Figure 3B: Overlays pH 7.0/7.8 with 1 M NaCl

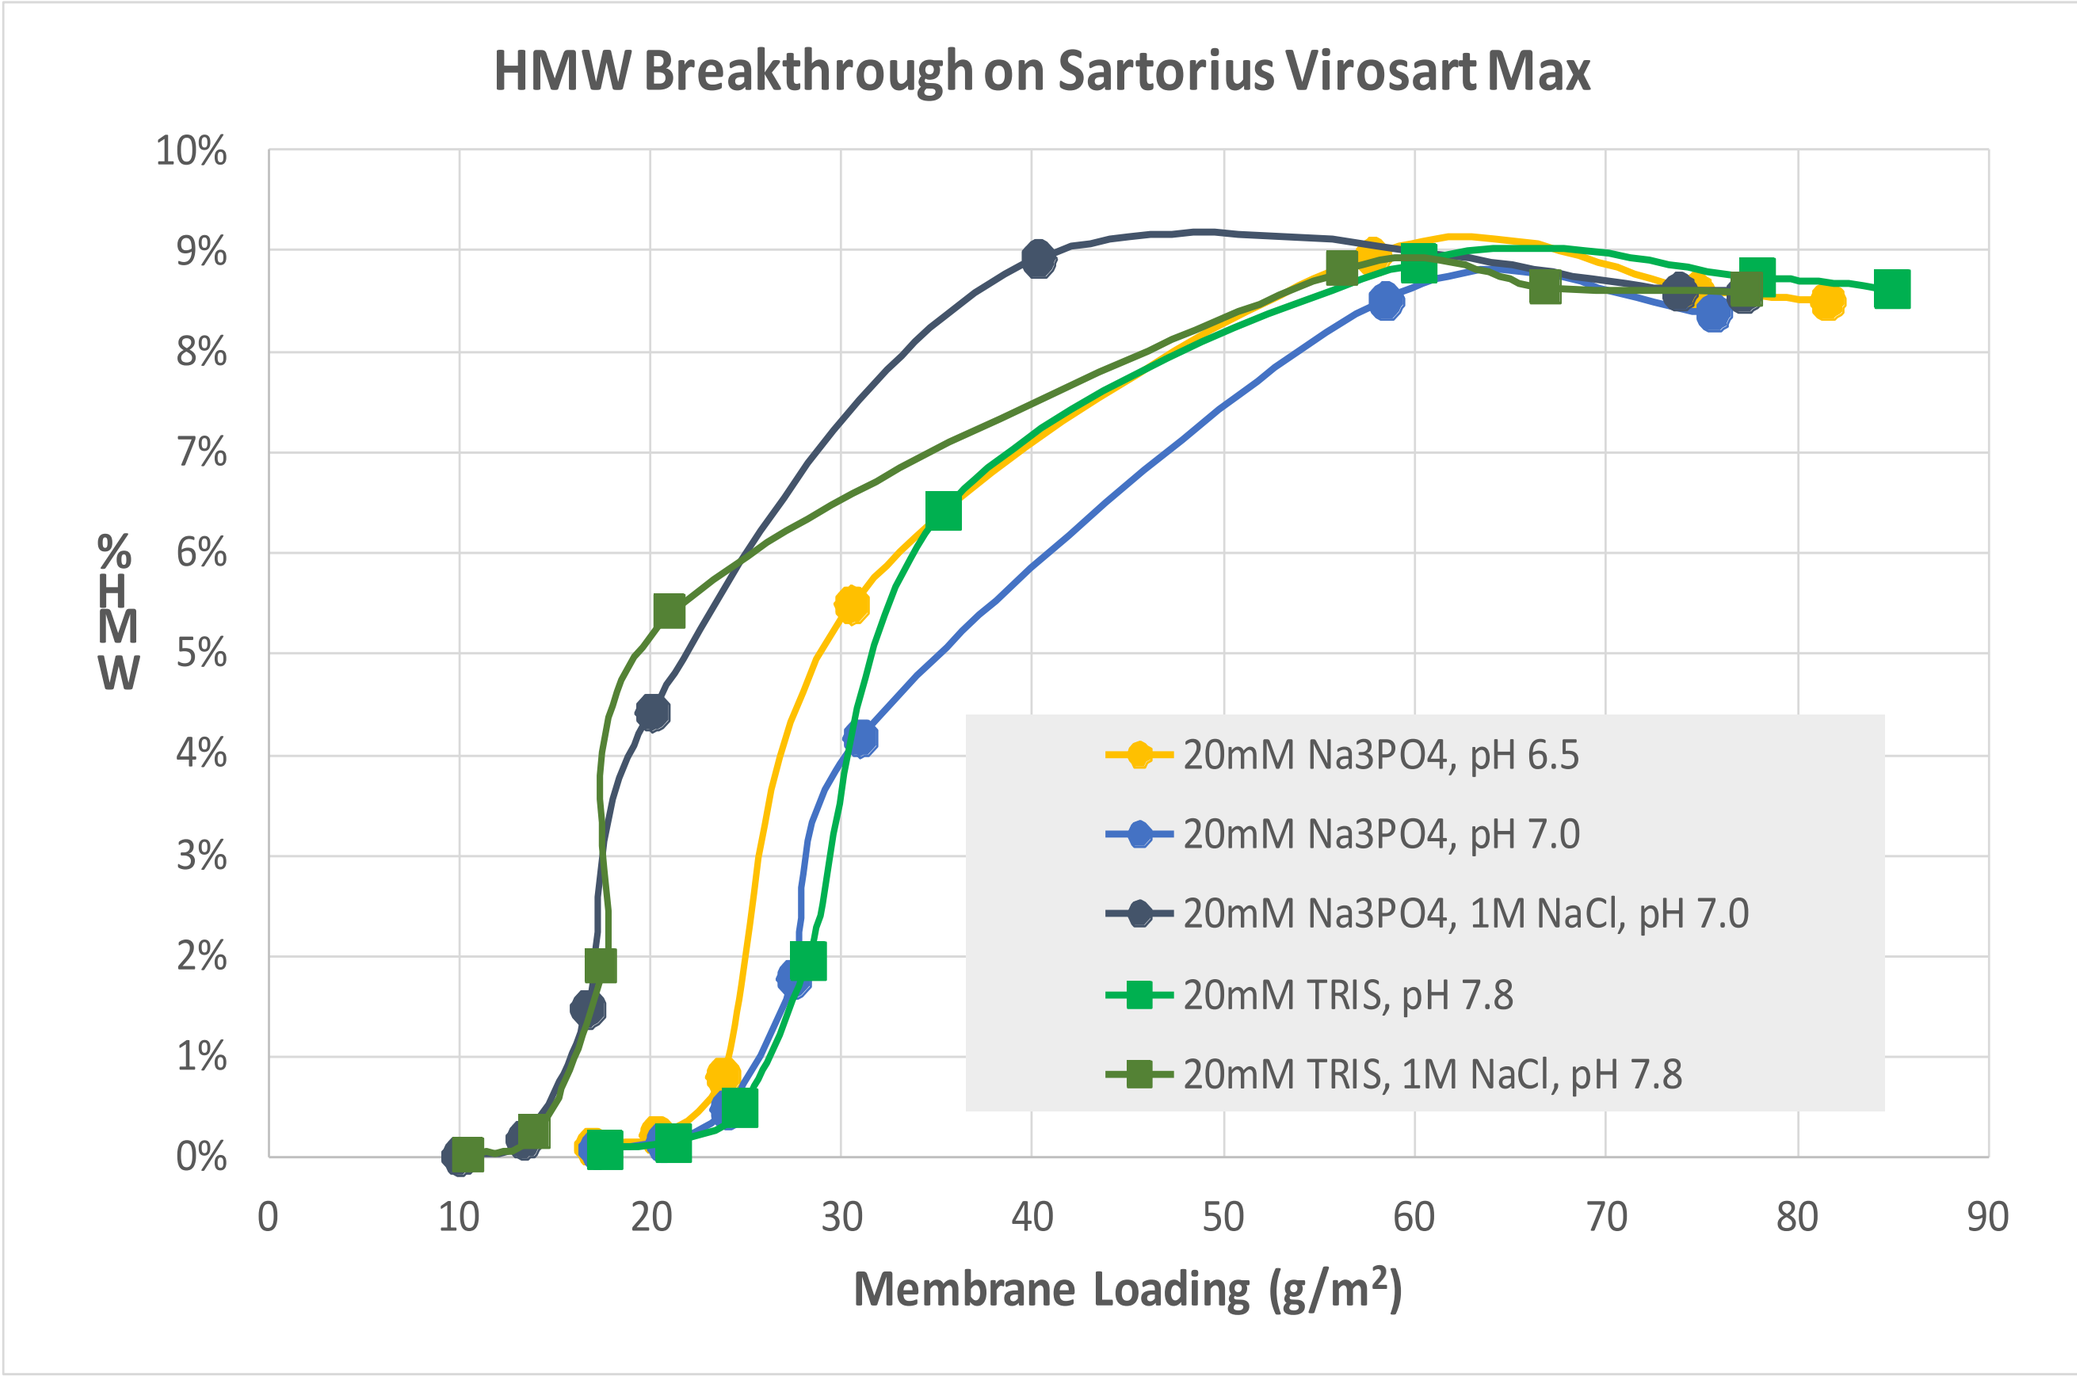
HMW Breakthrough on Sartorius Virosart Max
%HMW
Membrane Loading (g/m²)
20mM Na3PO4, pH 6.5
20mM Na3PO4, pH 7.0
20mM Na3PO4, 1M NaCl, pH 7.0
20mM TRIS, pH 7.8
20mM TRIS, 1M NaCl, pH 7.8

Figure 4: Breakthrough Curves on Virosart® Max Filter

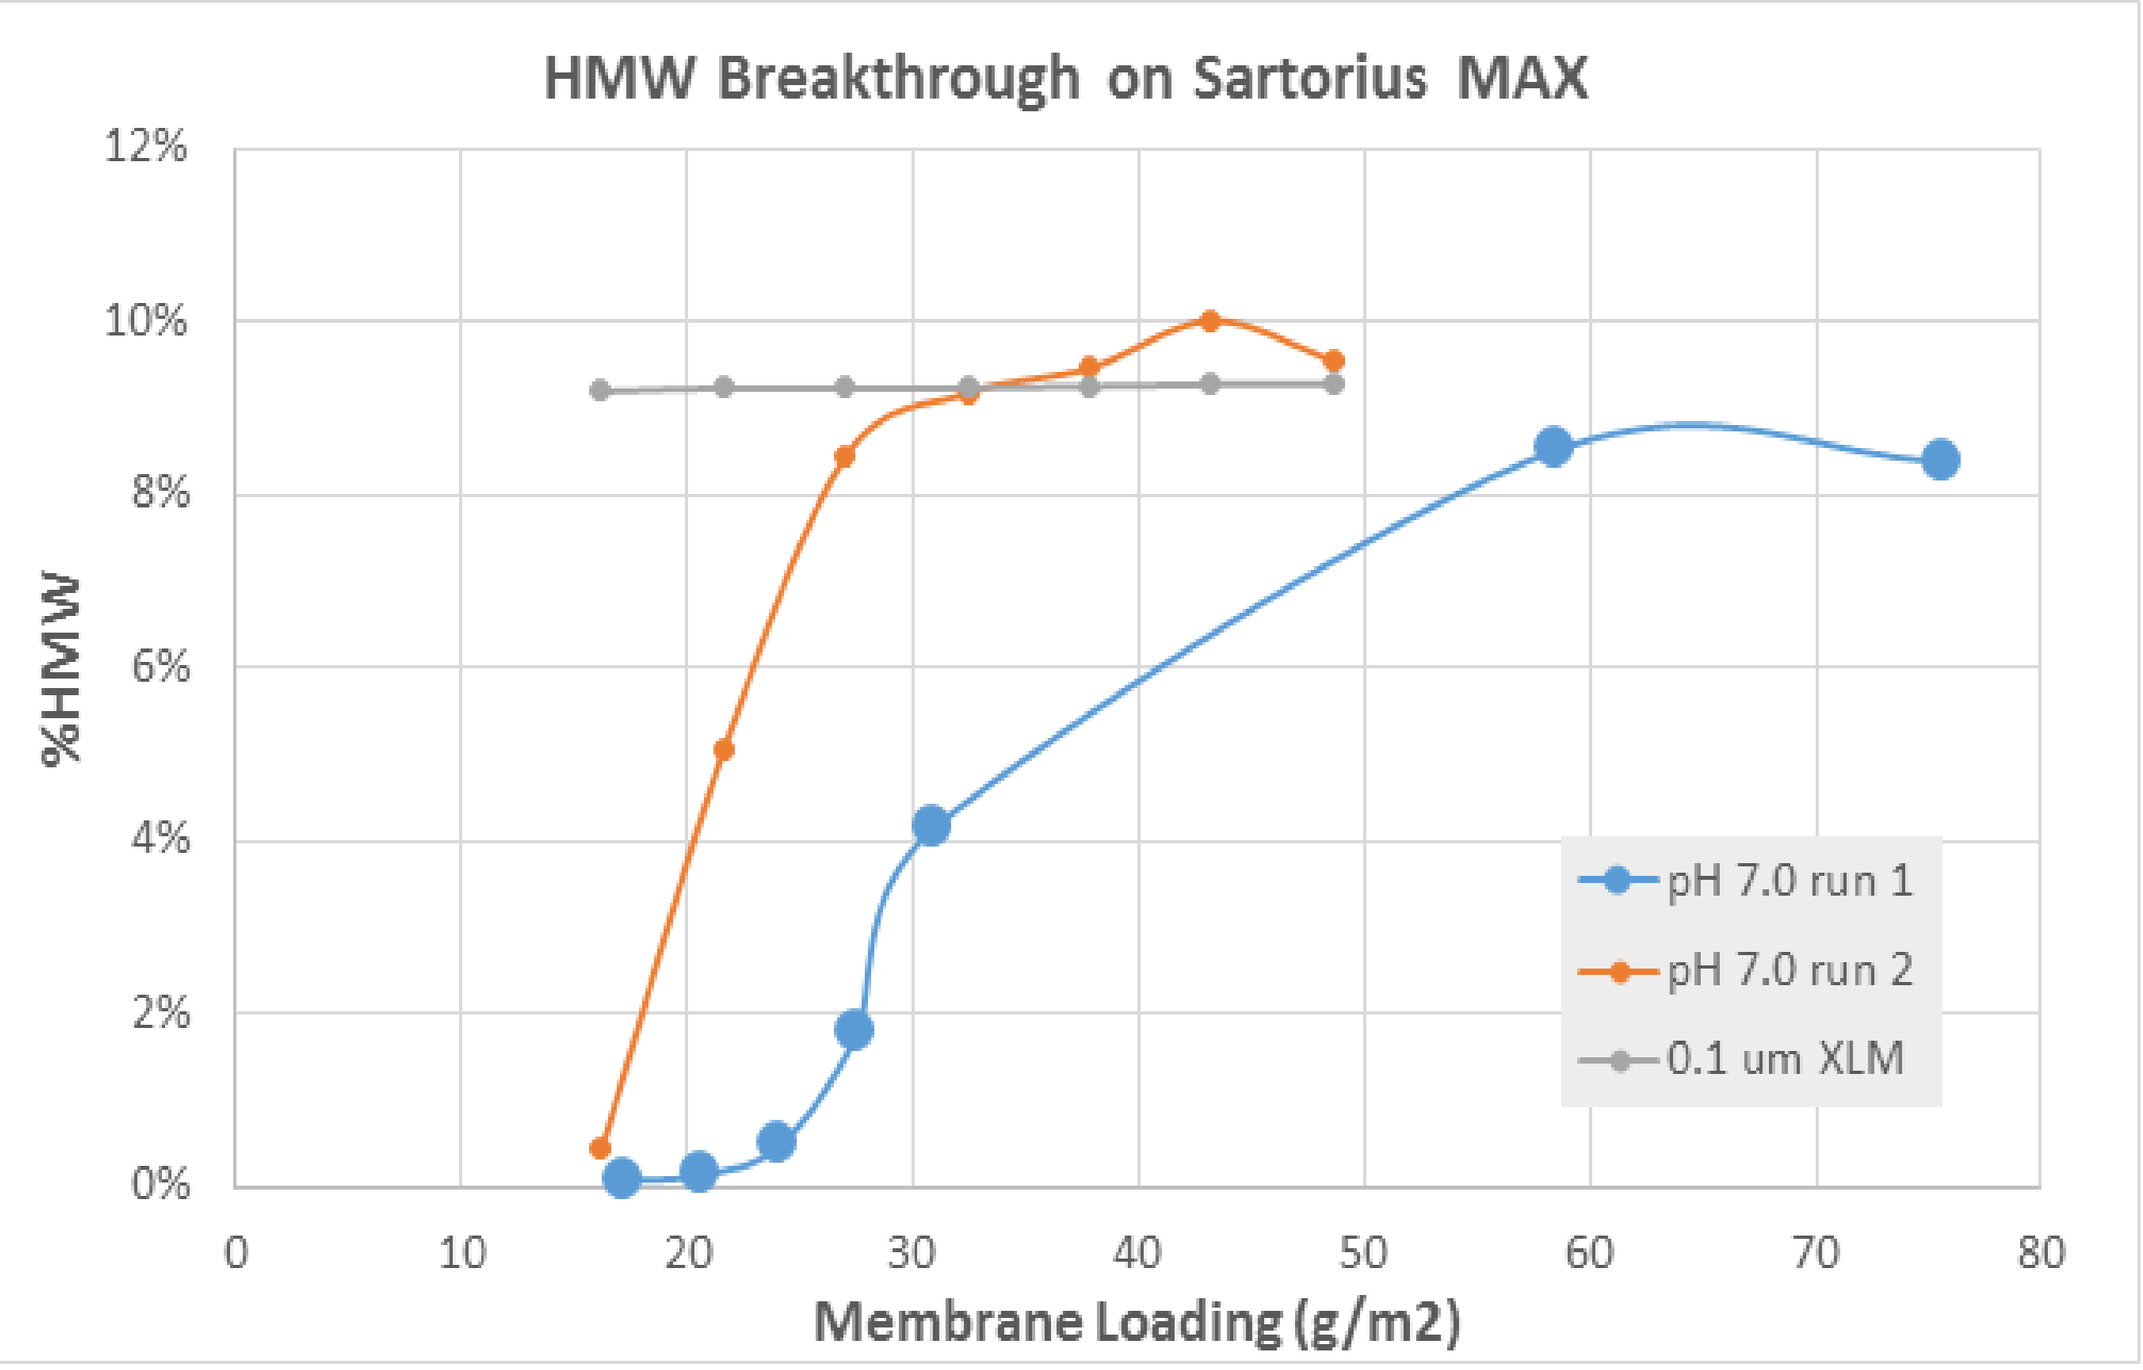
HMW Breakthrough on Sartorius MAX
%HMW
Membrane Loading (g/m2)
pH 7.0 run 1
pH 7.0 run 2
0.1 um XLM

Figure 5: Comparison of Virosart® Max and 0.1 µm Filter

Two distinct effects are observed with salt concentration and pH:

Salt Concentration

- By increasing salt concentration, the aggregate had earlier break through (over 6%) by the time 25 g/m^2 is reached.
- The low salt concentration increased performance of aggregate removal as expected since aggregation is often triggered by high salt concentration.

pH

- At pH 6.5, aggregate was observed breaking through faster than the pH 7.0 and 7.8
- At pH 7.0, the 6% aggregate level was reached at 40 g/m^2.
- At pH 7.8, the 6% aggregate level was reached at 35 g/m^2.

Conclusion

The results from this preliminary evaluation show reduction of aggregate (HMW species) using only the Virosart® Max pre-filter. No aggregate reduction was noted using the 0.1 µm filter. Both pH and salt concentration had significant impact on the overall aggregate breakthrough. Lower pH is less effective and increased salt concentrations proved detrimental to aggregate reduction. The bench scale model loaded 14.2 g/m^2 and the combined fractions recovered ~60% with reduction of aggregates (HMW) from 11.4% to <4.0%. As this is a method for a highly aggregated bi-specific antibody molecule, it is a worst case scenario and additional studies should be completed using other less-aggregated feed streams to verify the robustness of the filtration step.

View publication stats